Multicentre Open Trial Demonstrates Efficacy of Sublingual Immunotherapy in Canine Atopic Dermatitis

D. J. DeBoer,[1] M. Morris[2]

[1]School of Veterinary Medicine, University of Wisconsin, Madison, WI USA and [2]Allergychoices Inc., La Crosse, WI USA

ABSTRACT

Allergen-specific immunotherapy is commonly administered via the sublingual route (SLIT) in human atopic disease. There is renewed interest in SLIT for atopic dermatitis in man, especially with recent evidence that it may function by different mechanisms than does injection immunotherapy. A previous pilot study of SLIT in dogs sensitive to house dust mites provided evidence of clinical benefit and coincident immunologic changes. The present study evaluated the clinical efficacy of SLIT in a larger group of dogs. Nine veterinary dermatology specialty clinics enrolled a total of 217 dogs with atopic dermatitis in an open study on the efficacy of SLIT. All dogs received twice-daily administration of an escalating-dose, non-aqueous SLIT formulation devised according to individual tested sensitivities. The response of each patient after at least 6 months of SLIT was graded by the clinician according to four subjective response categories. Of 124 evaluable cases, 68 dogs (55%) were judged to have a good-to-excellent response to SLIT. Among these 124 dogs, 77 dogs that had received no previous immunotherapy had a response rate of 59%. The remaining dogs (n=47) had failed injection immunotherapy due to lack of efficacy, adverse reactions, or compliance difficulties. Of these injection failures, 23 dogs (49%) had a good-to-excellent response to SLIT. In this multicentre, open trial, we conclude that SLIT appears to be an effective treatment for canine atopic dermatitis, including in dogs that have failed injection immunotherapy.

BACKGROUND

Sublingual immunotherapy (SLIT) is allergen specific immunotherapy via administration of allergen extracts into the oral cavity, instead of by subcutaneous injection. SLIT is commonly used in Europe for allergic diseases in humans, but less so in the USA. Historically, there are conflicting reports of efficacy of SLIT. Widely-differing dosing protocols, allergen concentrations, intervals, vehicles, etc. may in part be responsible for the variation in results reported. Published efficacy studies are typically European, perhaps because SLIT administration is registered for human use in Europe, but not in North America. Recently however, increasing data and evidence- based reviews support the safety and efficacy of SLIT in human allergic disease.[1,2] Most studies of SLIT are in human atopic rhinitis and asthma, though studies do demonstrate its effectiveness in human atopic dermatitis (AD) as well.[3,4] We have reported the results of a pilot study of SLIT in canine AD, wherein ten dust mite-sensitive dogs were treated with SLIT for 6 months. Clinical improvement occurred in 8/10 dogs, and was accompanied by reduction in dust-mite specific IgE and increase in dust-mite specific IgG.[5,6]

OBJECTIVE

- **The purpose of the present study was to evaluate clinical benefit of SLIT treatment in a larger, more diverse group of dogs with AD, in a multicentre, open-label field study.**

MATERIALS AND METHODS

SLIT therapy was studied in an open-label, uncontrolled field study conducted at the Veterinary Medical Teaching Hospital (VMTH), University of Wisconsin-Madison and at eight other geographically-diverse, U.S. dermatology specialty clinics,[a] with a total of 18 veterinarians participating.

Patients. **All dogs entering the study were diagnosed with AD by board-certified dermatologists, and tested for allergic sensitivities by intradermal and/or serologic testing, as preferred by each participating doctor. Dogs were sensitive to multiple allergens, including dust, pollen, and/or mold components. Most dogs had not been treated previously with immunotherapy; some had failed previous treatment with subcutaneous immunotherapy ("allergy shots").**

Treatment Protocol. **SLIT therapy was initiated using a 3-vial set of escalating allergen concentration, prepared from glycerinated extracts in a proprietary vehicle, by a commercial supplier[b] in pump-type dispenser bottles. Owners were instructed on how to administer the treatment by hooking the dispenser tip over the lower teeth and dispensing solution into the oral cavity, under the tongue if possible, twice daily every day (Fig. 1). Concurrent medications as necessary to control symptoms and secondary infections were allowed initially, with the goal of tapering such additional medication and eventually discontinuing it if response to SLIT occurred.**

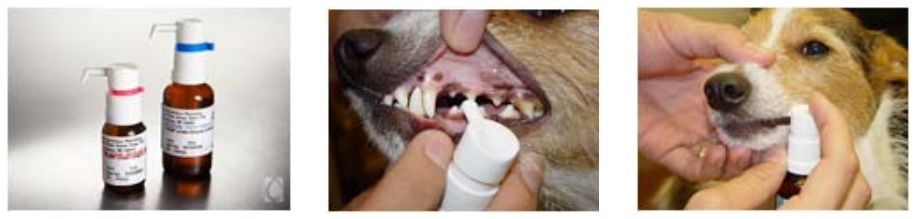

Figure 1: Administration of SLIT treatment.

Clinical Response Scoring. **A global clinical improvement scoring system ("Response Category") was used by each veterinarian to assess clinical response to SLIT (Table 1). The Response Category was based upon overall degree of control of the AD, along with the necessity for use of concurrent medications. After at least 6 months of SLIT therapy, the veterinarian assigned each patient to one of four Response Categories A thorough D, with D representing the best response. Categories "NF" or "ND" were assigned if response could not be determined due to lack of followup or concurrent medication use, respectively.**

Table 1: "Response Category" system for evaluation of patient clinical responses by the veterinarian.

RESPONSE CATEGORY	OVERALL DEGREE OF CONTROL	CONCURRENT MEDICATIONS	DESCRIPTION OF PATIENT RESPONSE
A	Disease control is not evident, or there is partial control.	Any combination (including "none")	SLIT, with or without other medications, is not producing improvement. OR, there is some control of the disease, but only by use of standard doses of concurrent medication, similar to what was used before institution of SLIT. Any control is felt likely to be due to the medication, rather than SLIT. SLIT clearly is not benefiting this patient. The owner does not wish to continue SLIT because it is not perceived as effective.
B	At least partial control of the disease is evident.	Anti-inflammatory drugs (steroids or CsA) are necessary for control, in standard doses. Other drugs may be used as necessary.	Though the disease is being controlled, anti-inflammatory medication is necessary for the control. However, there is a subjective feeling that addition of SLIT has produced slight incremental improvement or is allowing somewhat less concurrent medication use than previously. The veterinarian and owner are unsure of the value of SLIT, and are considering possibly stopping it.
C	Disease is under partial to good control.	Anti-inflammatory drugs (steroids or CsA) may be necessary for control, at reduced doses. Other drugs may be used as necessary.	Disease is under good control. Though anti-inflammatory medication may be necessary, it is at doses that are intermittent, or low, perhaps much lower than previously. OR, the dog is under partial, acceptable control on SLIT only without additional medications. SLIT has clearly produced some improvement, and the owner wishes to continue treatment.
D	Disease is under good control.	At the most, antihistamines, fatty acids, or antimicrobials may be necessary for control.	Disease is under good control. No additional medications may be necessary, or if used, additional medications are limited to antihistamines, fatty acids, or antimicrobials. SLIT has clearly produced marked improvement in this dog, and the owner is eager to continue treatment.
ND	Any	Any	Not determinable at this time. These pets are current on their recheck exams, but the response to SLIT is uncertain or cannot be determined because other medications are still being used in addition to the SLIT.
NF	Any	Any	Not determinable due to lack of followup, poor client compliance, early discontinuation for mechanical reasons, death due to unrelated cause, etc.

RESULTS

A total of 217 dogs were evaluated for their response to SLIT treatment (86 from the UW-VMTH and 131 from other practices). Of these dogs, 48 could not be evaluated due to lack of followup ("NF"). An additional 45 dogs were not evaluable at the time of data collection because concurrent medication had not yet been tapered ("ND"). In all, 124 of the patients had evaluable responses. Successful response to treatment was defined as either Response Category C or D (good-to-excellent; disease under control with SLIT with little or no additional concurrent medication needed). Using this definition, the overall successful response rate was 55% (Fig. 2a). Of 77 dogs that had not had previous immunotherapy, 59% responded to SLIT (Fig. 2b). In 47 dogs that had failed prior injection immunotherapy, the response rate was 49% (Fig. 2c).

Figure 2: Results of SLIT treatment.

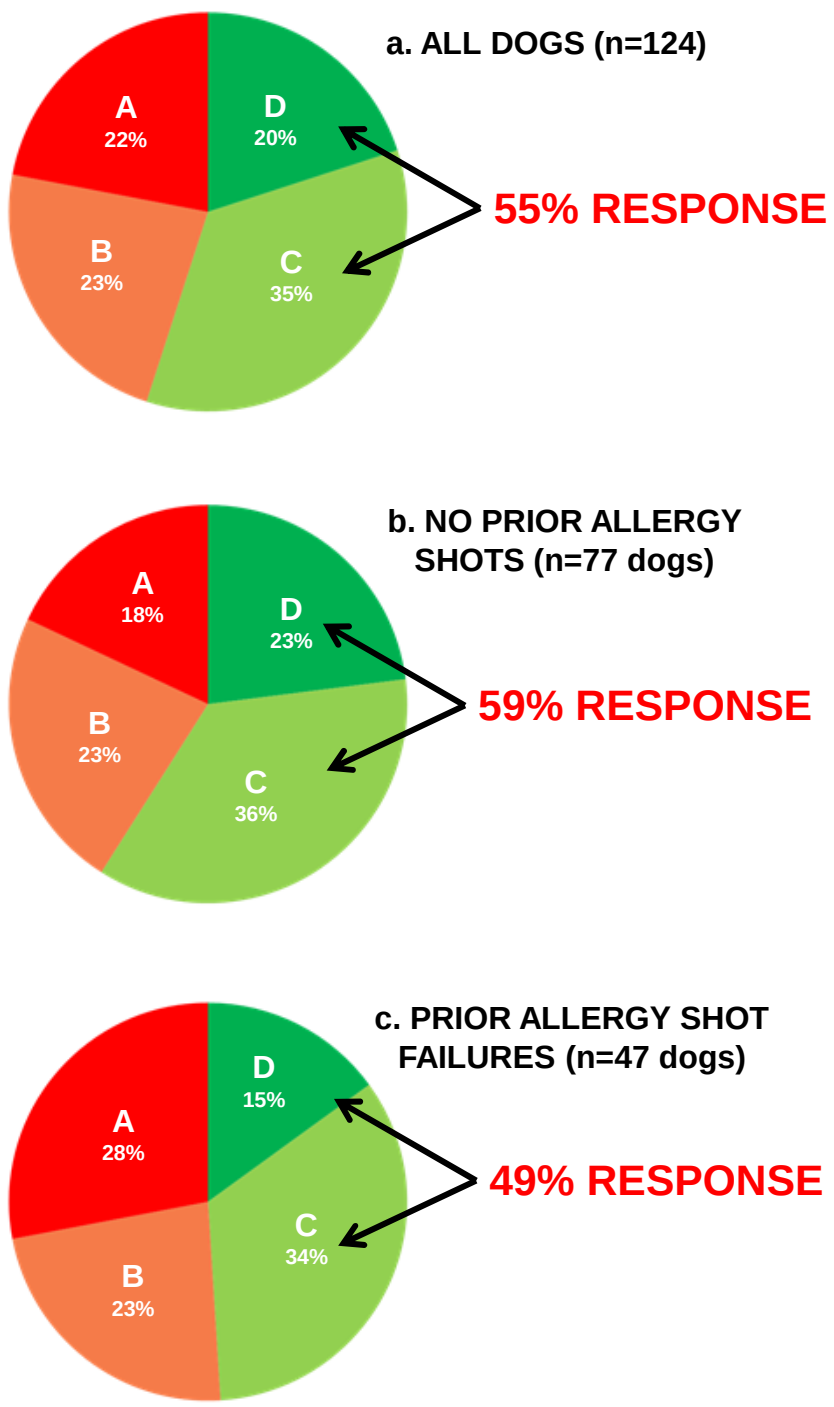
a. ALL DOGS (n=124)
A 22%
D 20%
B 23%
C 35%
55% RESPONSE
b. NO PRIOR ALLERGY SHOTS (n=77 dogs)
A 18%
D 23%
B 23%
C 36%
59% RESPONSE
c. PRIOR ALLERGY SHOT FAILURES (n=47 dogs)
A 28%
D 15%
C 34%
B 23%
49% RESPONSE

DISCUSSION

- **Data collected was subjective and empirical. Response rates are estimates, since a large number of patients were not evaluable for one reason or another.**
- **Successful SLIT treatment in allergy shot failures suggests the mechanism of action of SLIT in dogs may differ from that of injection immunotherapy, an observation that has an established basis in human immunologic studies.**
- **Some dogs had experienced anaphylaxis from allergy shots, and were safely treated with SLIT; this is also the case in human beings.**
- **We were impressed how many owners were pleased to not have to give their pets injections. SLIT may allow more owners to access immunotherapy, who would not have considered it previously.**

CONCLUSIONS

- **In this open field trial, sublingual immunotherapy (SLIT) was a successful treatment in 59% of evaluable patients who had not had previous immunotherapy. This approximates the response typically reported for subcutaneous immunotherapy.**
- **In addition, SLIT was a safe and successful treatment in 49% of evaluable patients who had failed previous allergy shot treatment**
- **Further studies are warranted including controlled trials and additional study of serologic changes occurring during treatment.**

REFERENCES AND FOOTNOTES

[a]The authors are grateful to the following veterinary clinics for participation: Dermatology Clinic for Animals (Las Vegas, NV); Upstate Veterinary Specialties (Latham, NY); McKeever Dermatology Clinics (Eden Prairie, MN); Veterinary Referral Center of Colorado (Englewood, CO); BluePearl Veterinary Partners (Tampa, FL); Veterinary Dermatology Center (Maitland, FL); Riverdale Veterinary Dermatology (Riverdale, NJ); College of Veterinary Medicine, Texas A&M University (College Station, TX).

[b]Allergychoices Inc., La Crosse, WI USA

[1]Bousquet PJ, Cox LS, Dunham SR, et al. Sublingual immunotherapy: World Allergy Organization Position Paper 2009. *World Allergy Organization Journal* 2:233-81, 2009

[2]Radulovic S, Wilson D, Calderon M, et al. Systemic reviews of sublingual immunotherapy (SLIT). *Allergy* 66:740-52, 2011.

[3]Cadario G, Galluccio AG, Pezza M, et al. Sublingual immunotherapy efficacy in patients with atopic dermatitis and house dust mite sensitivity: a prospective study. *Curr Med Res Opin* 23:2503-6, 2007.

[4]Pajno GB, Caminiti L, Vita D, et al. Sublingual immunotherapy in mite-sensitized children with atopic dermatitis: a randomized, double-blind, placebo-controlled study. *J Allergy Clin Immunol* 120:164-70, 2007.

[5]DeBoer DJ, Verbrugge M, Morris M. Pilot trial of sublingual immunotherapy in mite-sensitive atopic dogs (abstract). *Proceedings, North American Veterinary Dermatology Forum,* Portland, OR, April 2010.

[6]DeBoer DJ, Verbrugge M, Morris M. Changes in mite-specific IgE and IgG levels during sublingual immunotherapy (SLIT) in dust mite-sensitive dogs with atopic dermatitis (abstract). *Proceedings, European Society of Veterinary Dermatology/European College of Veterinary Dermatology Meeting,* Firenze, Italy, September 2010.

FUNDING and DISCLOSURES

Treatment materials were provided by Allergychoices, Inc. M. Morris is an owner of Allergychoices, Inc., and has licensed to Heska Corporation the exclusive rights to manufacture and distribute the sublingual immunotherapy formulation described in this study, under the trade name Allercept® Therapy Drops. D. DeBoer is currently a consultant for Heska Corporation, but was not during the time this study was conducted.